Differential dependency of CD19 chimeric antigen receptor (CAR)-T (TX019) and CD19- (CAR) natural killer (NK) cells (NKX019) on CD58 loss in Acute Lymphoblastic Leukemia (ALL)

Carmel T. Chan, Max Zhang, Nikki Kimura, Jessica Sood, Damie Juat, Tao Geng, and James Trager
Preclinical Research, Nkarta Therapeutics, South Francisco, CA 94040, United States

Introduction

- Acute lymphoblastic leukemia (ALL) has a 5-year survival rate of around 30-35% [1]. Current treatment for ALL includes chemotherapy, stem cell transplantation, and immunotherapy.
- NKX019 is an investigational CD19-targeting chimeric antigen receptor (CAR) natural killer (NK) cell therapy with engineered persistence for treating B cell malignancies. NKX019 exhibits more rapid cytotoxicity, lower production of cytokines associated with cytokine release syndrome, and detects target cells expressing lower CD19 levels than CD19-directed CAR T cells [2]. The safety and clinical activity of NKX019 is being evaluated in a Phase I clinical study [NCT05020678].
- CD58 is a co-stimulatory receptor that activates T cells and NK cells via its interaction with CD2 [3]. CD58 loss/mutation is associated with poor responses to CD19 CAR-T therapies and reduced survival in clinical and preclinical models of blood cancers [4,5].

Methods

A. NKX019 and TX019 cells were generated from the healthy donor-derived peripheral blood mononuclear cells as previously described [2]

B. Generation of Nalm6/CD19KO and Nalm6/CD58KO cells:
- ❖ Nalm6/CD19 and Nalm6/CD58 knock-out (KO) clones were derived from Nalm6/Luc2.eGFP/WT cells via CRISPR/Cas9. Cell-surface expression of CD19 or CD58, and CD2 binding were measured by flow cytometry.

C. *In vitro* cytotoxicity: Bright-Glo™ and Incucyte® assays:
- ❖ NKX019 and TX019 effector cells were incubated with target cells (Nalm6/WT, Nalm6/CD19KO, and Nalm6/CD58KO cells) at different effector to target (E:T) ratios in triplicates for 24 hours. EC_{50} values for cytotoxicity were determined by Bright Glo assay™ (Promega).
- ❖ Longitudinal killing efficacies of NKX019 and TX019 in Nalm6/WT, Nalm6/CD19KO, and Nalm6/CD58KO cells were determined by Incucyte® at different E:T ratios in triplicates. NKX019 and TX019 were rechallenged with additional target cells at 72 hours, followed by scanning for another 72 hours.

D. In vivo growth inhibition of Nalm6/WT, Nalm6/CD19KO, Nalm6/CD58KO tumor cells by NKX019 and TX019:
- ❖ All animal procedures were conducted in accordance with locally approved Animal Care and Use Protocols. Nalm6/WT, Nalm6/CD19KO, and Nalm6/CD58KO cells were inoculated into NSG mice prior to injection of 10E6 NKX019 (Days 0, 7, 14) or 1.2E6 TX019 (Day 0). Mice injected with 5% Flexbumin served as the negative controls. 5 mice were used for each treatment cohort. Tumor burden was determined by longitudinal bioluminescence imaging (BLI) using IVIS Spectrum (Perkin Elmer).

Data Analysis

All data are expressed as means +/- standard error of means (S.E.M.). Flow cytometry data analyses were performed using FlowJo™. GraphPad was used for determination of a) EC_{50} values for the BrightGlo using a 4-parmater logistic curve fitting; b) EC_{50} shift; and c) 2-way ANOVA for statistical comparisons between different groups. Non-parametric t-tests were used to compare statistical differences between different cohorts for the *in vivo* study.

Results

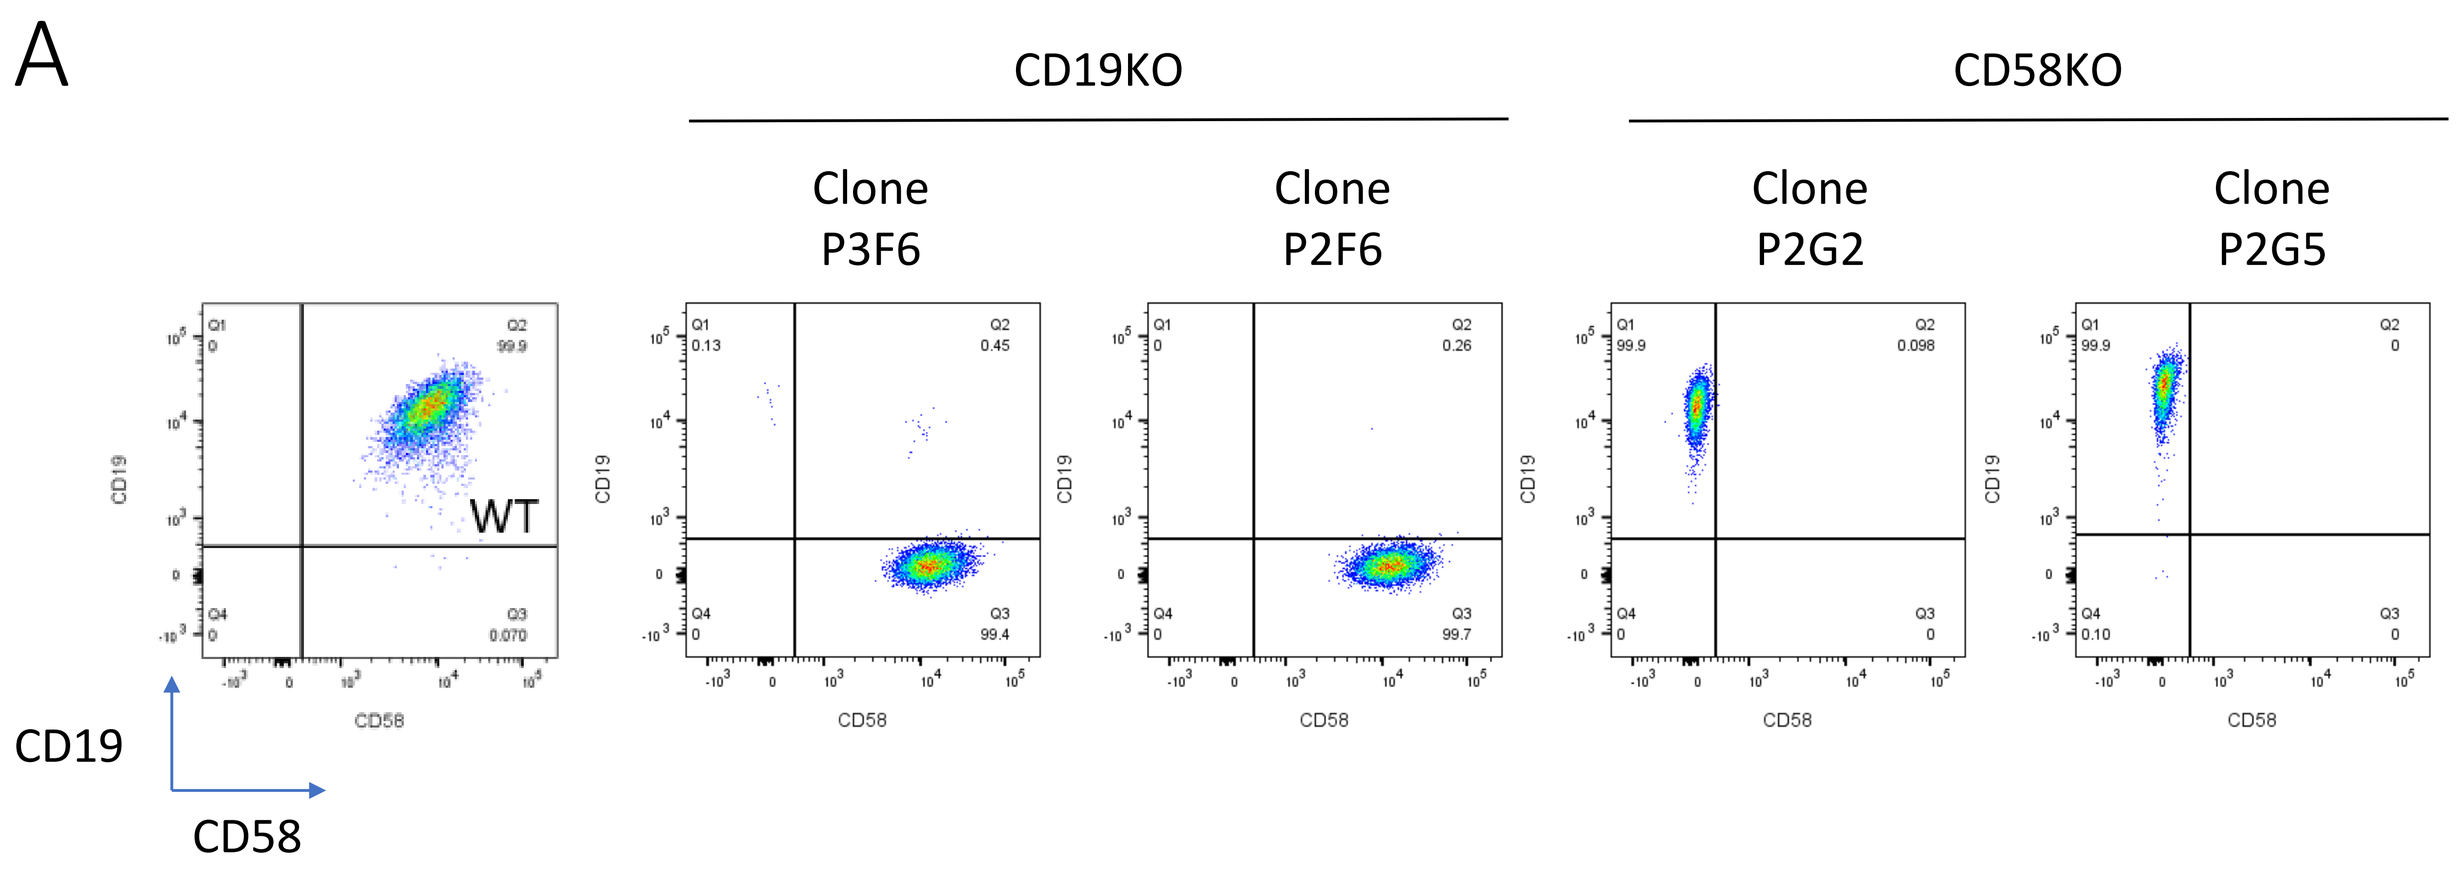
A
CD19KO
CD58KO
Clone P3F6
Clone P2F6
Clone P2G2
Clone P2G5
WT
CD19
CD58

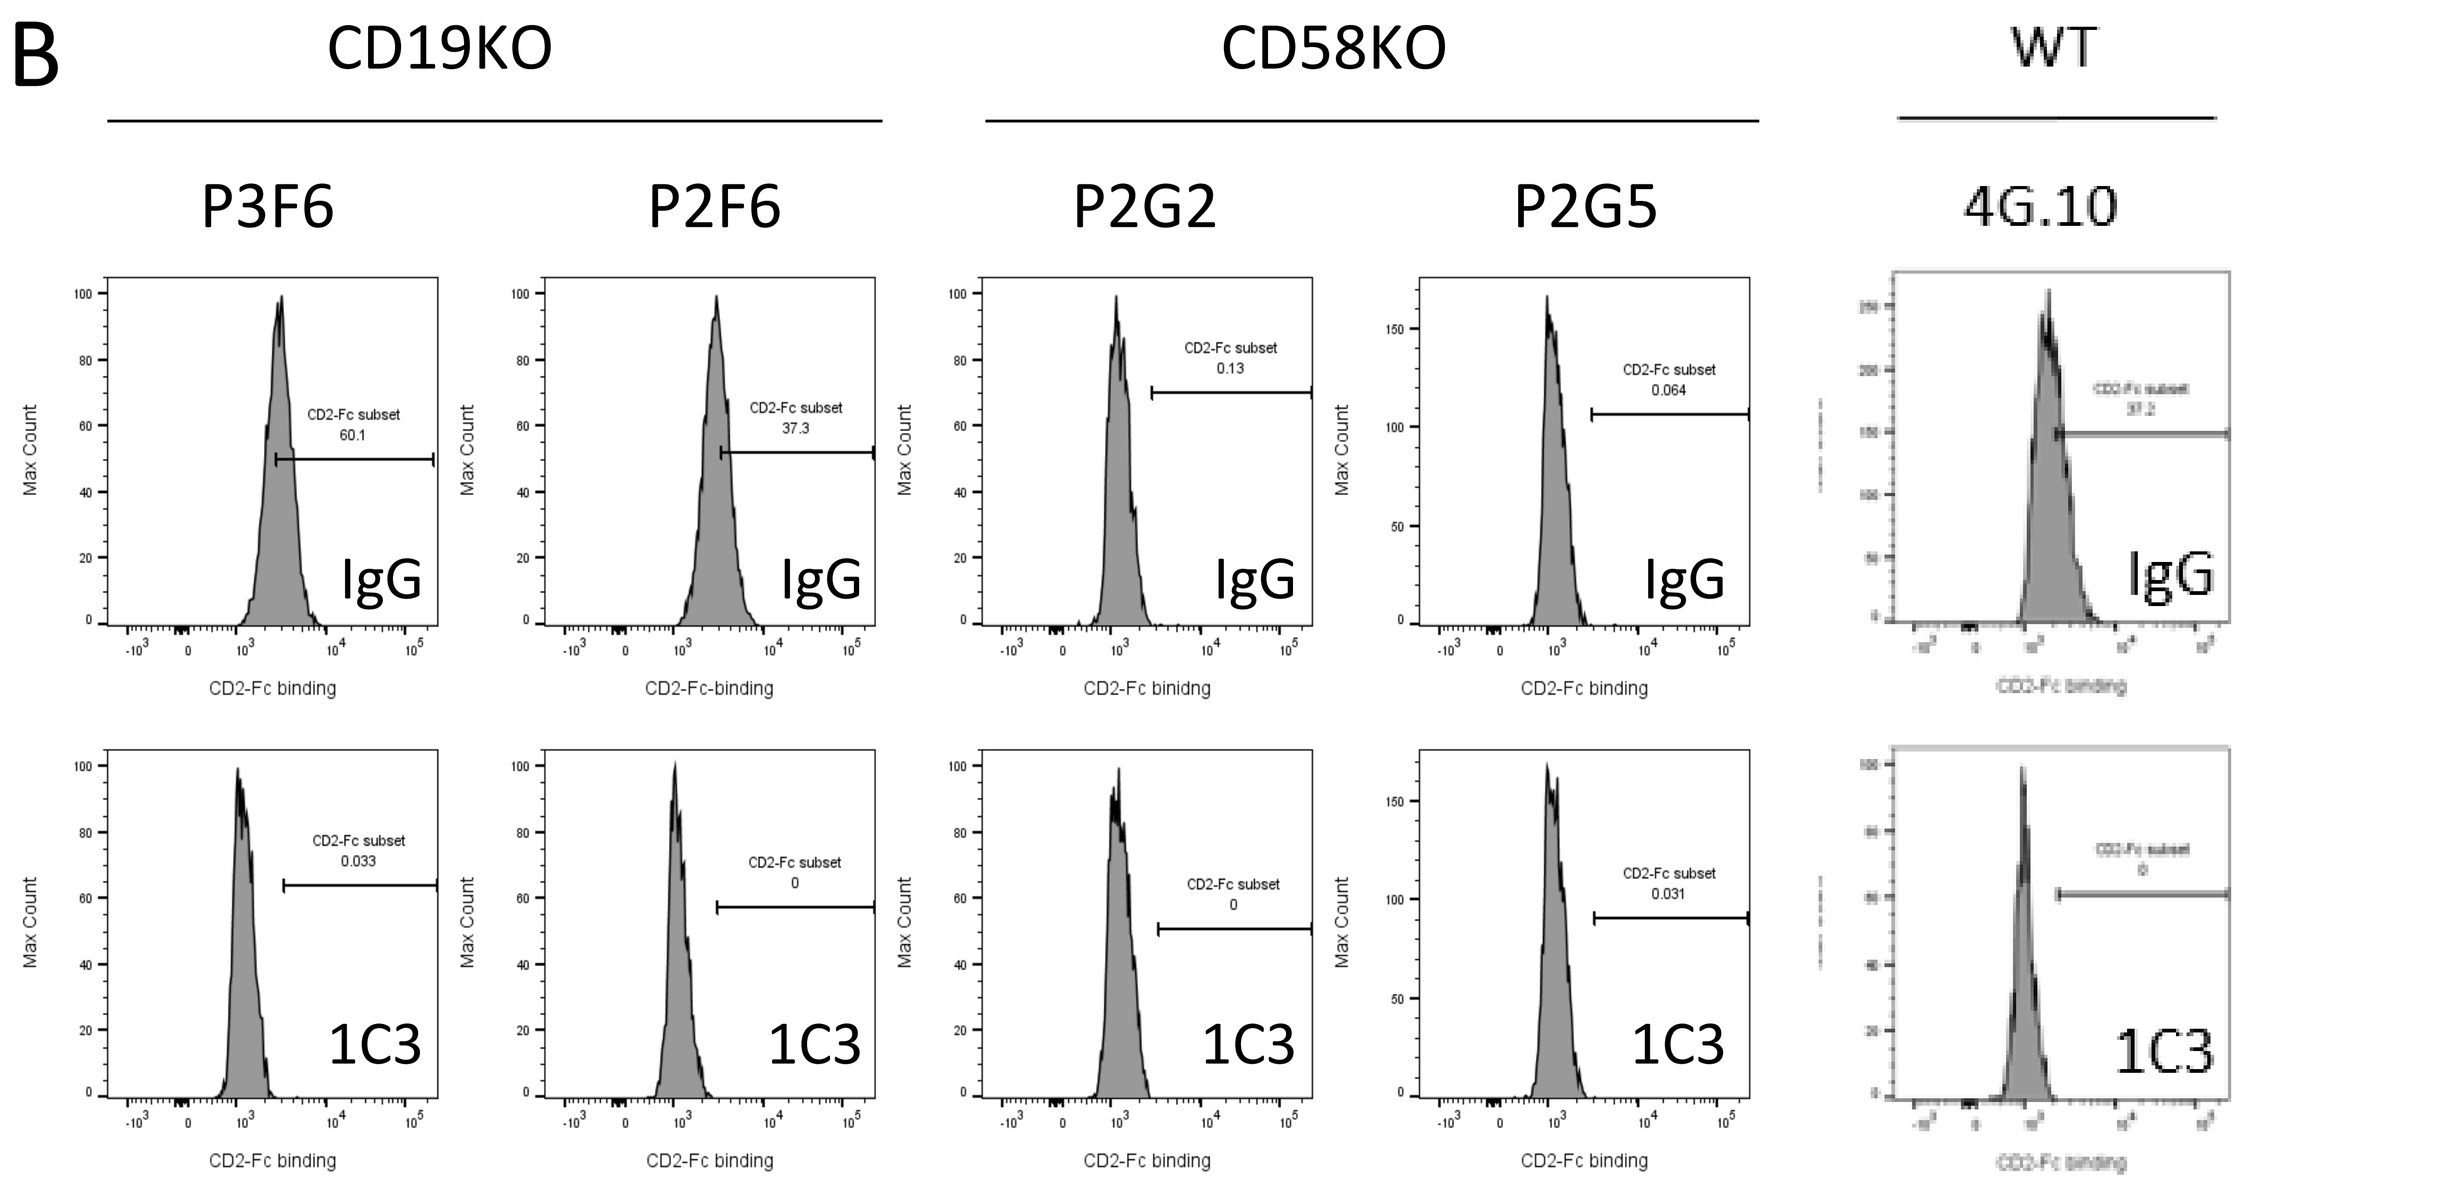
B
CD19KO
CD58KO
WT
P3F6
P2F6
P2G2
P2G5
4G.10
IgG
1C3

Figure 1: Characterization of Nalm6/CD19KO and Nalm6/CD58KO clones.

A. Cell-surface expression of CD19 and CD58 was absent in CD19KO and CD58KO clones, respectively.

B. Binding to CD2-Fc was abolished in CD58KO cells, but not in WT and CD19KO cells that express CD58. WT, CD19KO, and CD58KO cells were incubated with normal mouse IgG or CD58 blocking Ab 1C3, prior to incubation with CD2-Fc-biotin or IgG-Fc-biotin. The presence of bound CD2-Fc-biotin was detected by staining with PE-anti-biotin.

Figure 2: NKX019 was more potent than TX019 against Nalm6 cells with abrogated CD58 expression

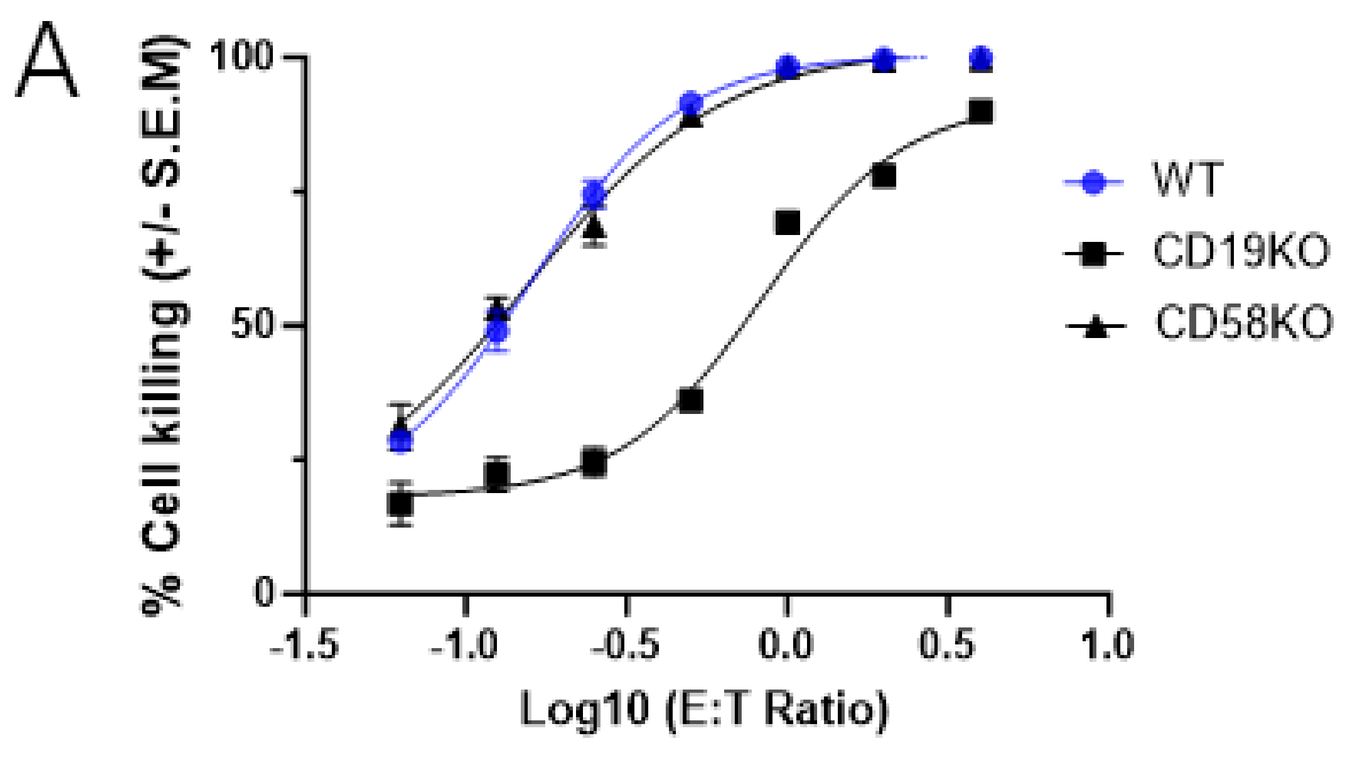
A
% Cell killing (+/- S.E.M)
Log10 (E:T Ratio)
WT
CD19KO
CD58KO

	WT	CD19KO	CD58KO
EC_{50}	0.1593	0.8346	0.1542
R^2	0.9896	0.9777	0.9771

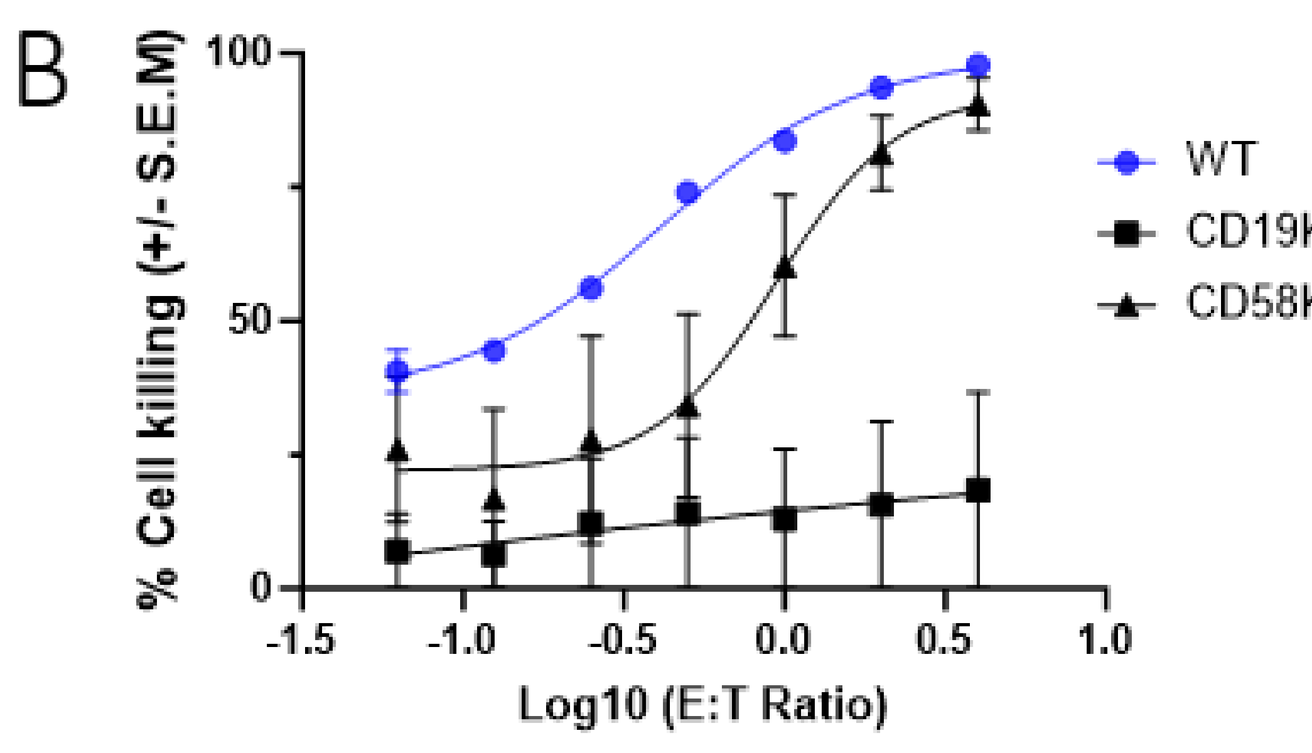
B
% Cell killing (+/- S.E.M)
Log10 (E:T Ratio)
WT
CD19KO
CD58KO

	WT	CD19KO	CD58KO
EC_{50}	0.4149	N.D.	0.9443
R^2	0.983	N.D.	0.6419

A. NKX019 was effective in killing of WT cells, but its efficacy was significantly diminished in CD19KO cells (p < 0.001). The killing efficacy of NKX019 for CD58KO cells was similar to that of WT cells.

B. TX019 was effective in killing of WT cells, but its efficacy was sharply diminished in CD19KO cells (p <0.0001). The killing efficacy of TX019 was in the also significantly reduced in CD58KO cells (p <0.001).

Figure 4: Unlike TX019, the *In vivo* efficacy of NKX019 was not significantly affected by CD58 loss

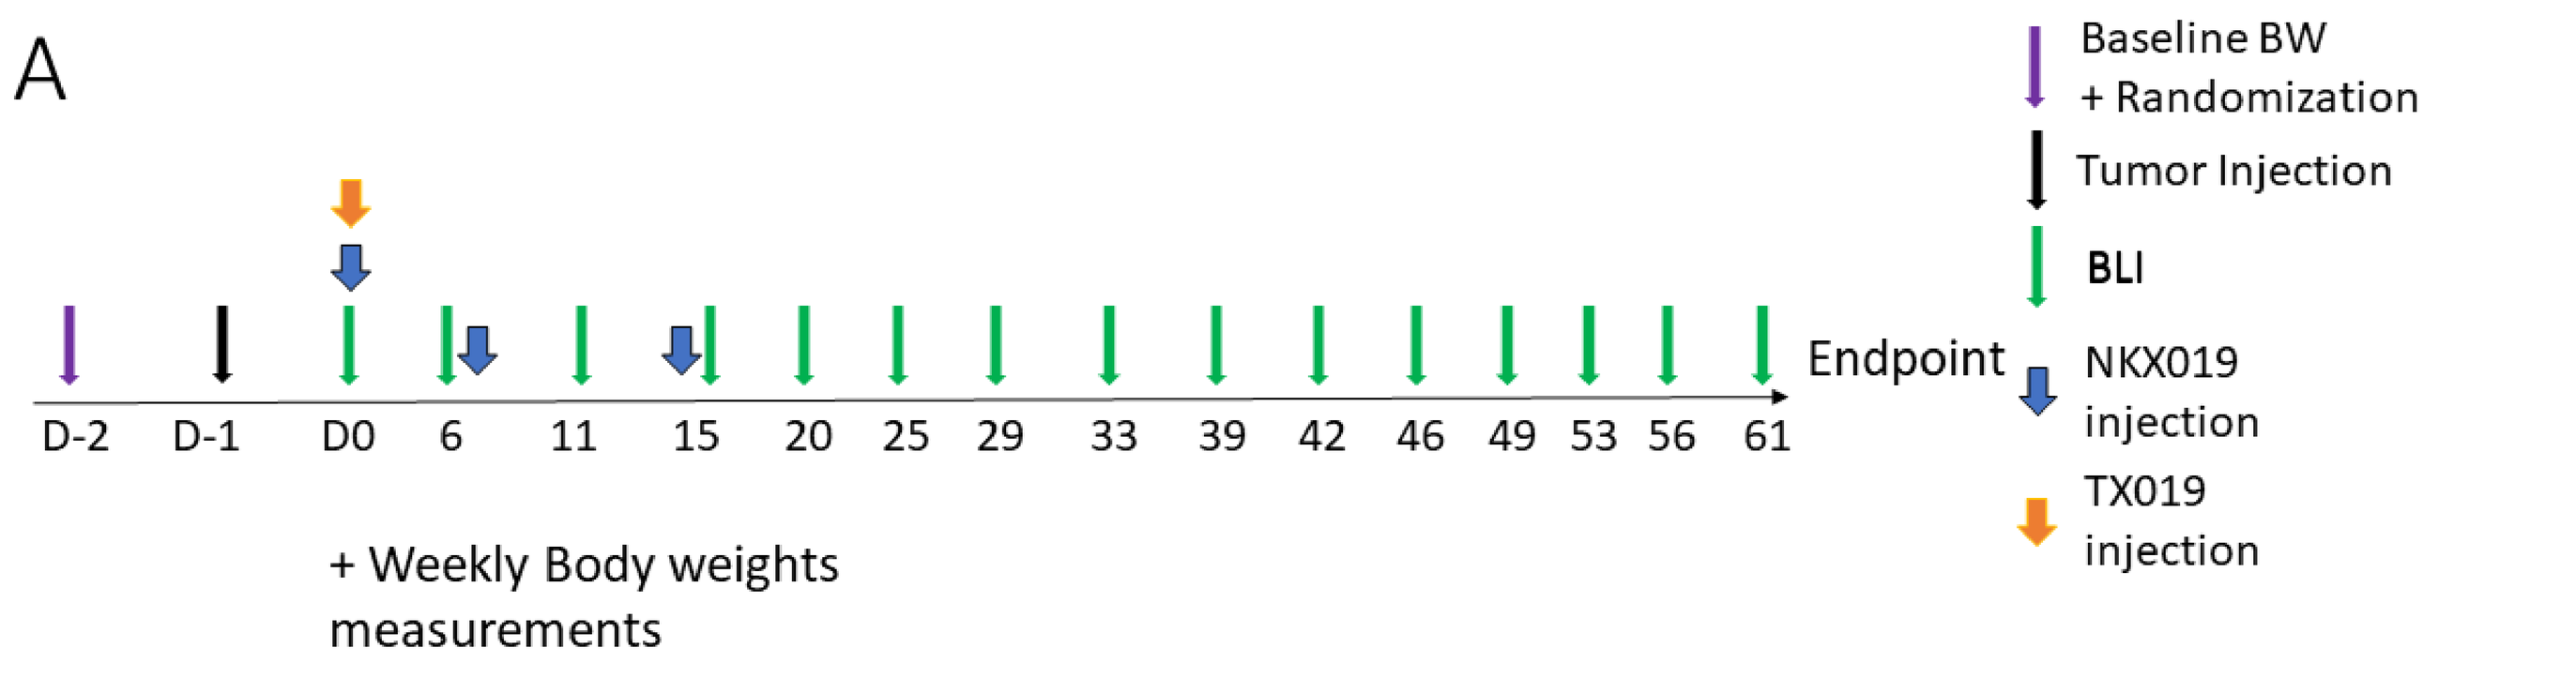
A
D-2
D-1
D0
6
11
15
20
25
29
33
39
42
46
49
53
56
61
Endpoint
+ Weekly Body weights measurements
Baseline BW + Randomization
Tumor Injection
BLI
NKX019 injection
TX019 injection

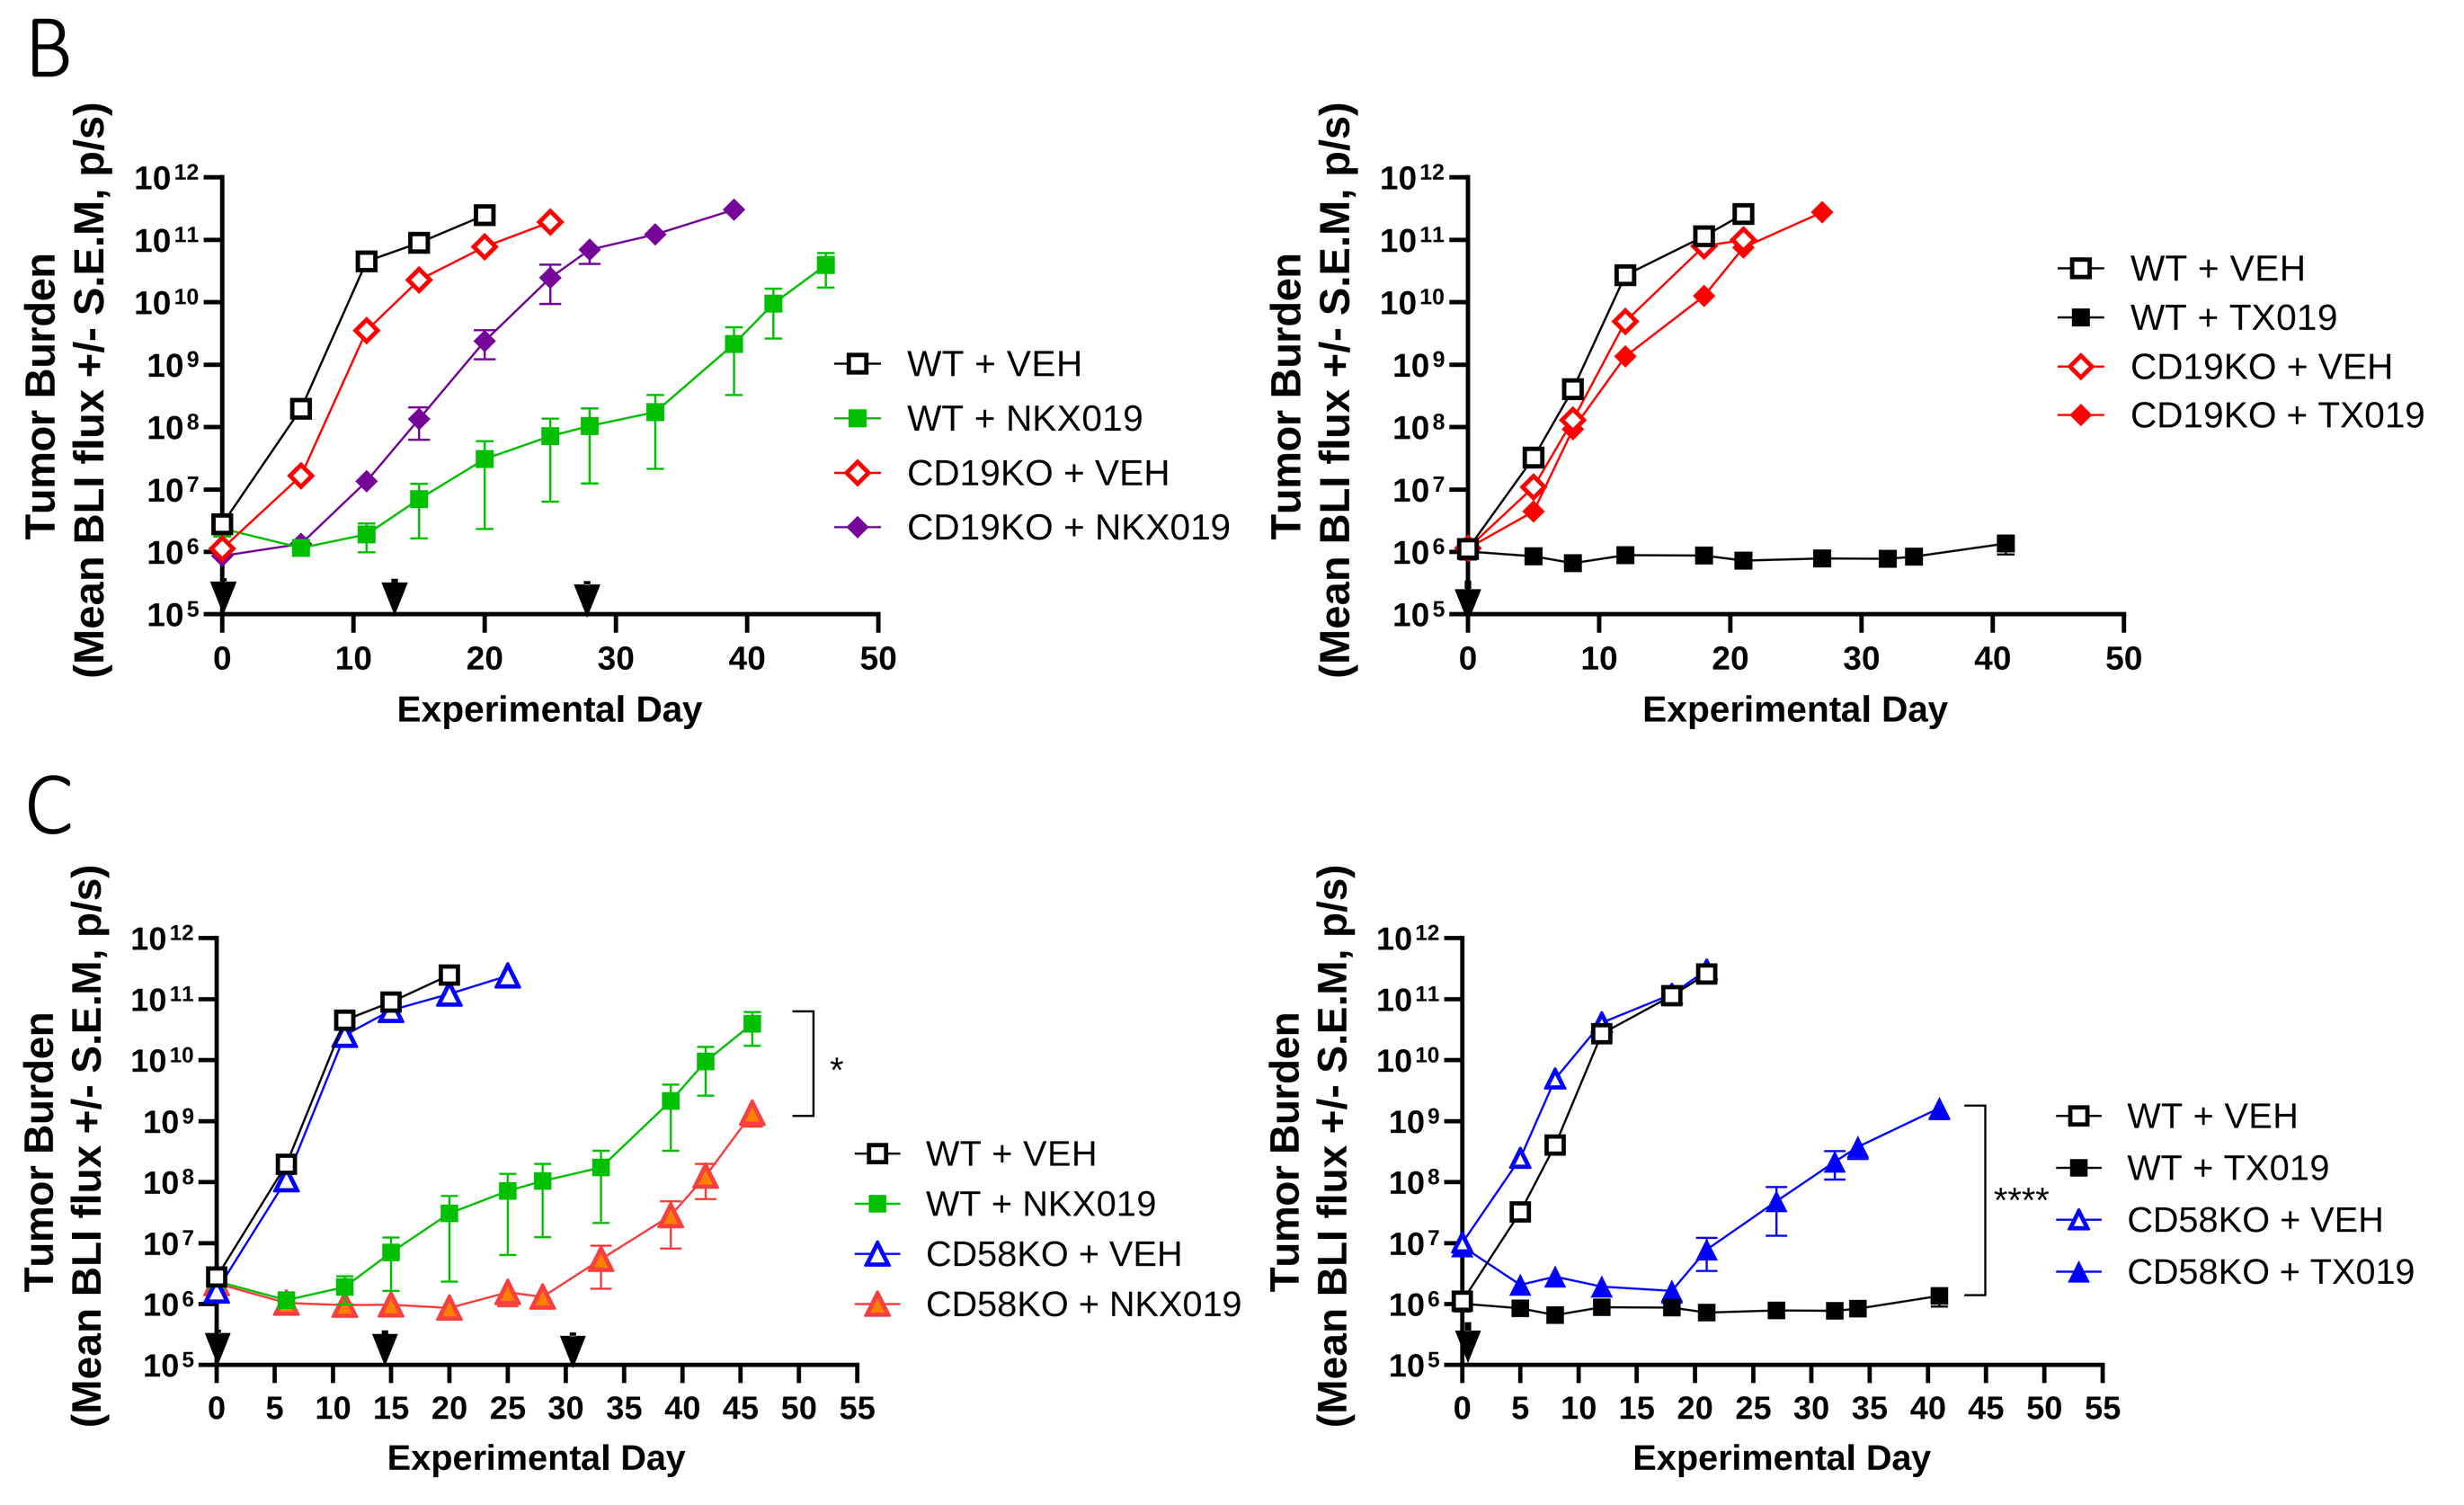
B
C
Tumor Burden (Mean BLI flux +/- S.E.M, p/s)
Experimental Day
WT + VEH
WT + NKX019
CD19KO + VEH
CD19KO + NKX019
WT + TX019
CD19KO + TX019
CD58KO + VEH
CD58KO + NKX019
CD58KO + TX019
*

A. In vivo NKX019 and TX019 treatment scheme for WT, CD19KO, CD58KO cells inoculated in NSG mice.

B. NKX019 and TX019 were more efficacious in growth inhibition in WT cells, compared to that of CD19KO cells. Black arrows denote the treatment days.

C. While NKX019 had similar efficacy in mice inoculated with WT and CD58KO cells, TX019 was less effective in *in vivo* growth inhibition of CD58KO cells. (*, p < 0.05; ****, p < 0.001)

Figure 3: Incucyte assays show that NKX019 had similar *in vitro* potency and persistency against WT and CD58KO cells, whereas TX019 potency and persistency was reduced in CD58KO cells compared to WT cells.

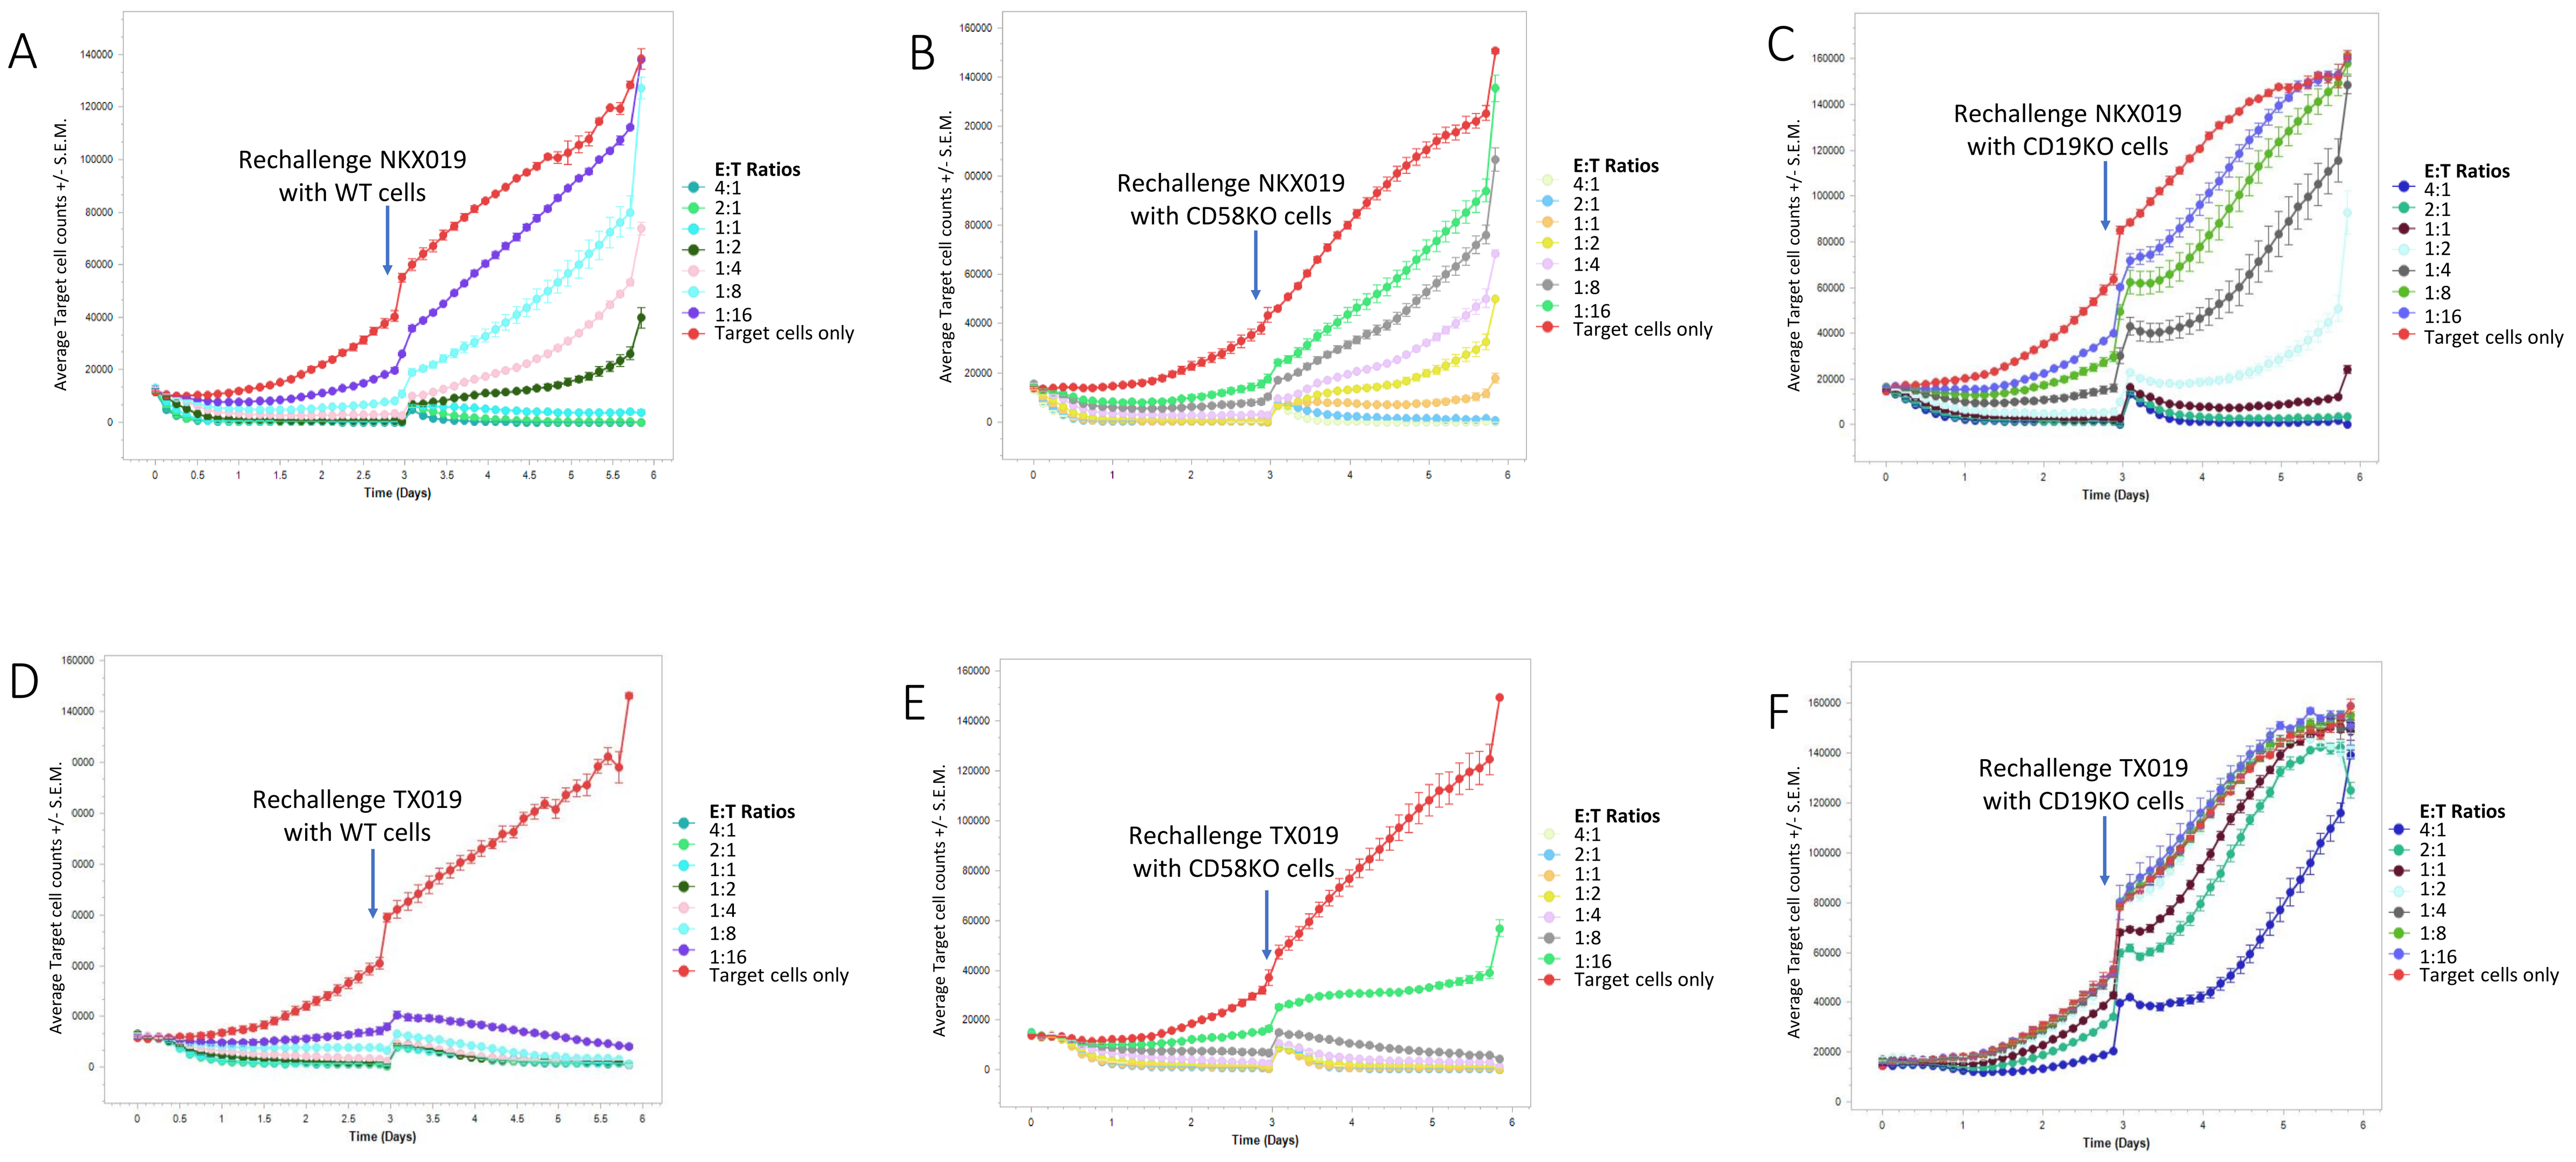
A
B
C
D
E
F
Rechallenge NKX019 with WT cells
Rechallenge NKX019 with CD58KO cells
Rechallenge NKX019 with CD19KO cells
Rechallenge TX019 with WT cells
Rechallenge TX019 with CD58KO cells
Rechallenge TX019 with CD19KO cells
Average Target cell counts +/- S.E.M.
Time (Days)
E:T Ratios
4:1
2:1
1:1
1:2
1:4
1:8
1:16
Target cells only

- NKX019 cytotoxicity was only modestly sensitive to CD58 loss (B). NKX019 cytotoxicity and resistance to rechallenge was attenuated when measured against CD19KO cells (C), compared to its activity against WT cells (A).
- CD58 loss led to decreased potency and persistency of TX019 (E). TX019 and resistance to rechallenge was attenuated in CD19KO cells (F), compared to its activity against WT cells (D).

Conclusions

- ❖ *In vitro* efficacy of NKX019 and TX019 was dependent on CD19 expression in target cells
- ❖ Loss of CD58 did not impact the *in vitro* efficacy of NKX019
- ❖ *In vitro* efficacy of TX019 was dependent on the presence of CD58 in target cells
- ❖ *In vivo* efficacy of NKX019 was not significantly affected by CD58 loss in target cells
- ❖ *In vivo* efficacy of TX019 was more dependent on the presence CD58 on the target cells, compared to that of NKX019
- ❖ **NKX019 may have an advantage over CD19 CAR-T in treatment of CD19-positive indications with CD58 loss/mutation(s)**

References

1. Recent Advances in Managing Acute Lymphoblastic Leukemia. *American Society of Clinical Oncology Educational Book* 40 (May 18, 2020) 330-342.
2. Morisot N, Wadsworth S, Davis T, *et al*. Preclinical evaluation of NKX019, a CD19-targeting CAR NK cell. J Immuno Therapy Cancer. 2020;8(suppl 3):A78
3. Zhang, Y. *et al*. CD58 immunobiology at a glance. Front Immunol (2021). 12: 705260
4. Majzner *et al*. CD58 aberrations limit durable responses to CD19 CAR in large B cell lymphoma patients treated with Axicabtagene Ciloleucel but can be overcome through novel CAR engineering. Blood (2020) 136 (Supplement 1): 53–54.
5. Yan, X *et al*. CD58 loss in tumor cells confers functional impairment of CAR T cells. Blood Adv (2022) 6 (22): 5844–5856.

Contact

James Trager, PhD
jtrager@nkartatx.com
www.nkartatx.com